This research was funded by:	This research was funded by Achieve Life Sciences.		
Please add "yes" or "no" to each table cell. If "yes", please turn cell background color to yellow.	Tobacco industry	E-cigarette & nicotine product industry (excluding pharma)	Pharma industry
The work being presented has received funding or other means of support from any of the following sources:	No	No	Yes
Any of the authors have received funding (including consultancy) from any of the following sources in the past 5 years:	No	Yes	Yes
Off-label medication uses discussed:	Cytisinicline is a partial agonist/antagonist with selective binding affinity for nicotine receptors and is under investigation for the treatment of nicotine dependence for smoking cessation in adults.		
Industry funding to the investigator in the last 5 years; list all:	MR and ML-A are employees and own stock/stock options in Achieve Life Sciences. CJ was an employee at Achieve Life Sciences at the time the current analysis was performed and owns stock/stock options in the company.		

Cytisinicline in adult smokers: Post-trial survey from ORCA-OL

Mark Rubinstein,[1] Matthew Linley-Adams,[1] Cindy Jacobs[1]*

*[1]Achieve Life Sciences, Seattle, WA, USA; *Employed by Achieve Life Sciences when the current analysis was performed*

Poster: 1-127

Introduction

- Cytisinicline is a plant-based alkaloid and partial agonist at α4β2 nicotinic acetylcholine receptors[1]
- A novel cytisinicline treatment regimen is currently under regulatory review, consisting of a 3 mg tablet administered 3 times daily (TID) for 6 or 12 weeks, as evaluated in the phase 3 ORCA-2 and ORCA-3 trials[2,3]
- In the randomized, placebo-controlled phase 3 ORCA-2 and ORCA-3 trials, cytisinicline was more effective than placebo in achieving continuous smoking abstinence in adults, with a favorable safety profile[2,3]
- In the randomized, placebo-controlled phase 2 ORCA-V1 trial, cytisinicline was more effective than placebo in achieving continuous abstinence from electronic (e)-cigarettes in adults, with a favorable safety profile[1]
- ORCA-OL is a multicenter, open-label (OL) phase 3 study of cytisinicline 3 mg TID for up to 52 weeks in adults who previously took part in the phase 2 (ORCA-V1) or phase 3 (ORCA-2 or ORCA-3) cytisinicline trials[1–4]
- Participants in ORCA-OL were daily cigarette smokers and/or users of daily nicotine-containing e-cigarettes, who were motivated to make another quit attempt[4]

Objective

To report the findings of a voluntary post-trial participant survey of cytisinicline use during the ORCA-OL trial[4]

Figure 1. Study design

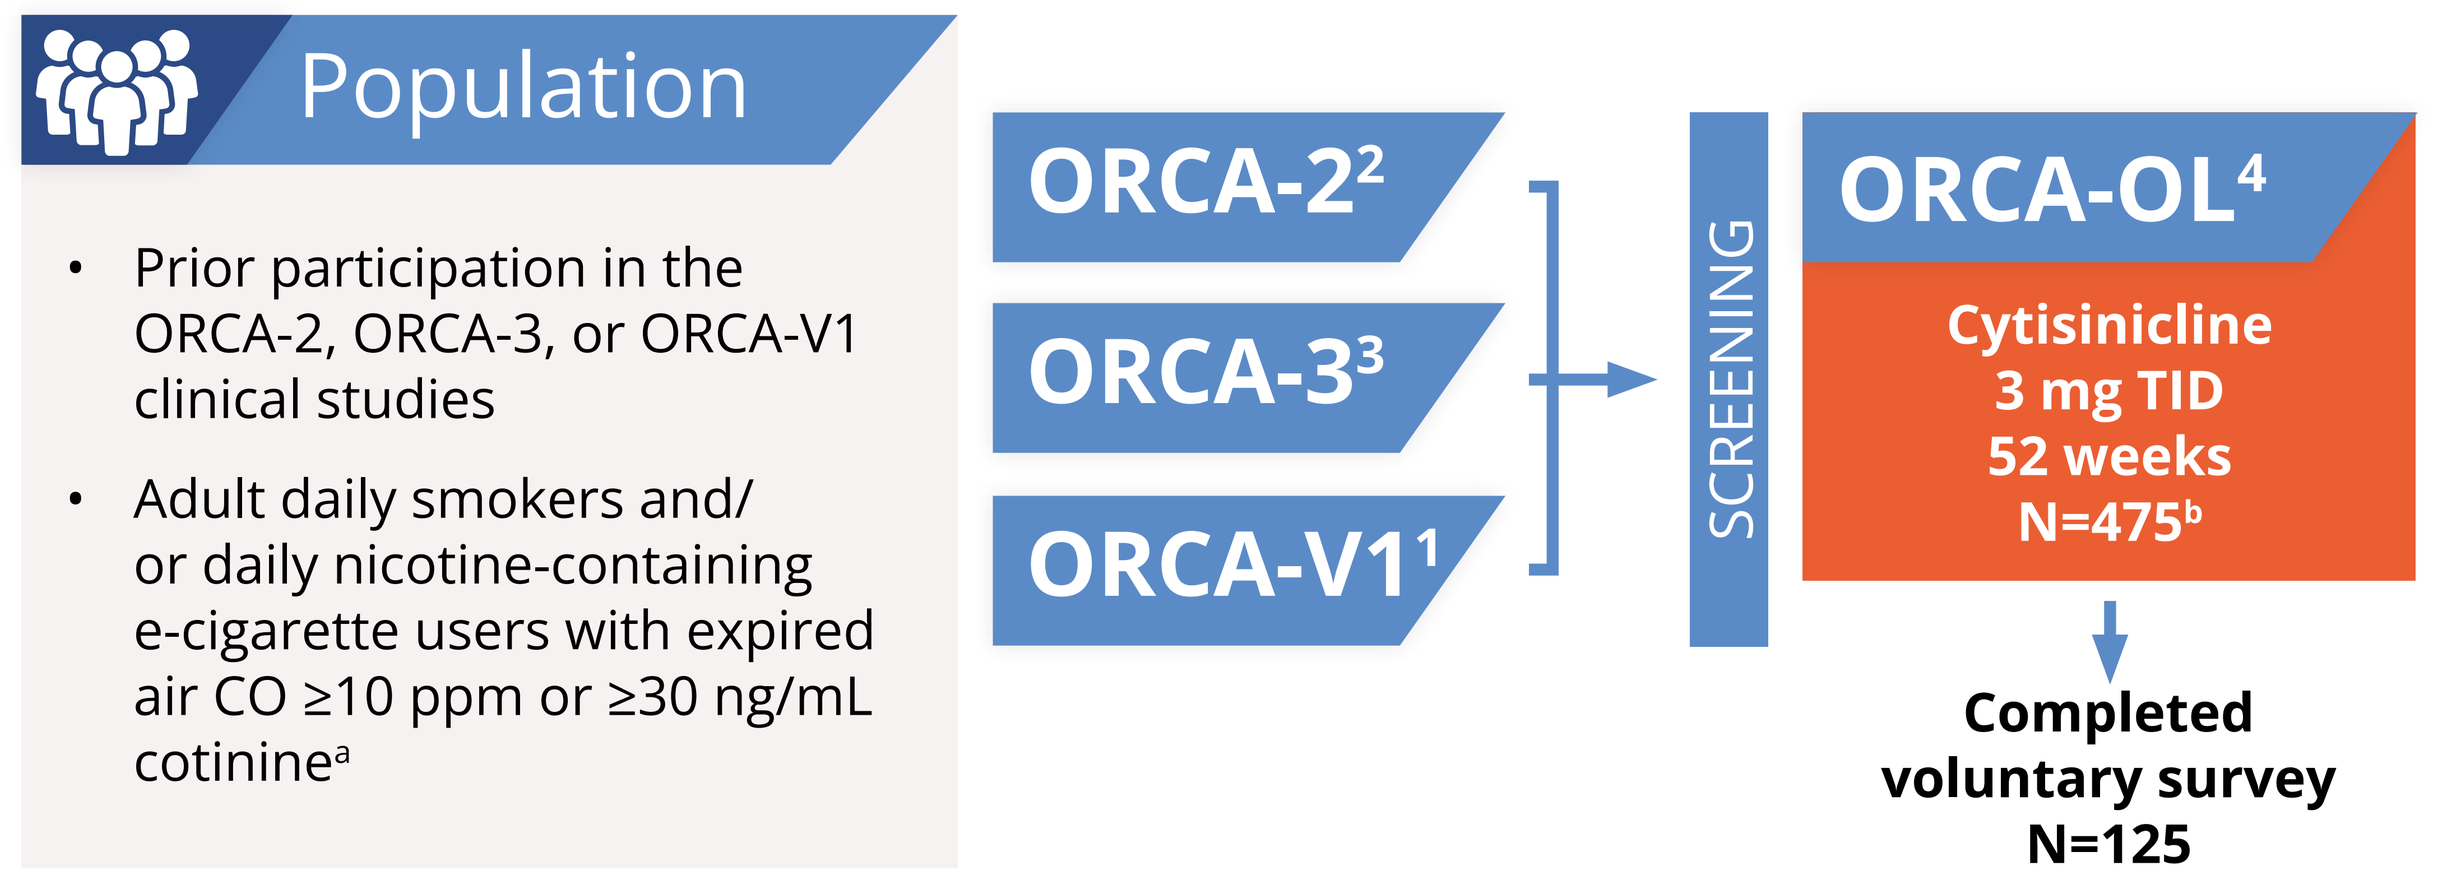

Population
Prior participation in the ORCA-2, ORCA-3, or ORCA-V1 clinical studies
Adult daily smokers and/or daily nicotine-containing e-cigarette users with expired air CO ≥10 ppm or ≥30 ng/mL cotinine[a]
ORCA-2[2]
ORCA-3[3]
ORCA-V1[1]
SCREENING
ORCA-OL[4]
Cytisinicline
3 mg TID
52 weeks
N=475[b]
Completed voluntary survey
N=125

[a]Using a point-of-care cotinine oral fluid screening device if self-reporting as users of nicotine-containing e-cigarettes; [b]Treated population. CO, carbon monoxide; e-cigarettes, electronic cigarettes; OL, open label; ppm, parts per million; TID, three times daily.

Materials and methods

- The ORCA-OL study took place at 29 sites across the USA
- Participants were invited to complete a voluntary, post-trial, 15-minute online questionnaire
- After entering their study subject ID, participants completed a structured post-trial survey consisting of closed- and open-ended questions

Results

Participants

- Of 475 participants in the ORCA-OL study, 125 completed the voluntary post-trial survey related to cytisinicline use **(Table 1)**

Table 1. Participants' main source of nicotine at the start of the survey

When I started this study, my main source of nicotine was:	Participants (N=125)
Cigarettes alone, n (%)	104 (83)
Nicotine e-cigarettes/vapes, n (%)	16 (13)
Both cigarettes and nicotine e-cigarettes/vapes, n (%)	5 (4)

Treatment expectation:
Participants were optimistic about their ability to quit during ORCA-OL, with over half (55%) expecting to quit and 21% expecting to reduce their smoking and/or vaping

Figure 2. Survey results

A. Motivation for trial participation and quitting smoking/vaping

Please select the reasons that you decided to participate in the ORCA-OL trial and to quit smoking or vaping[a]

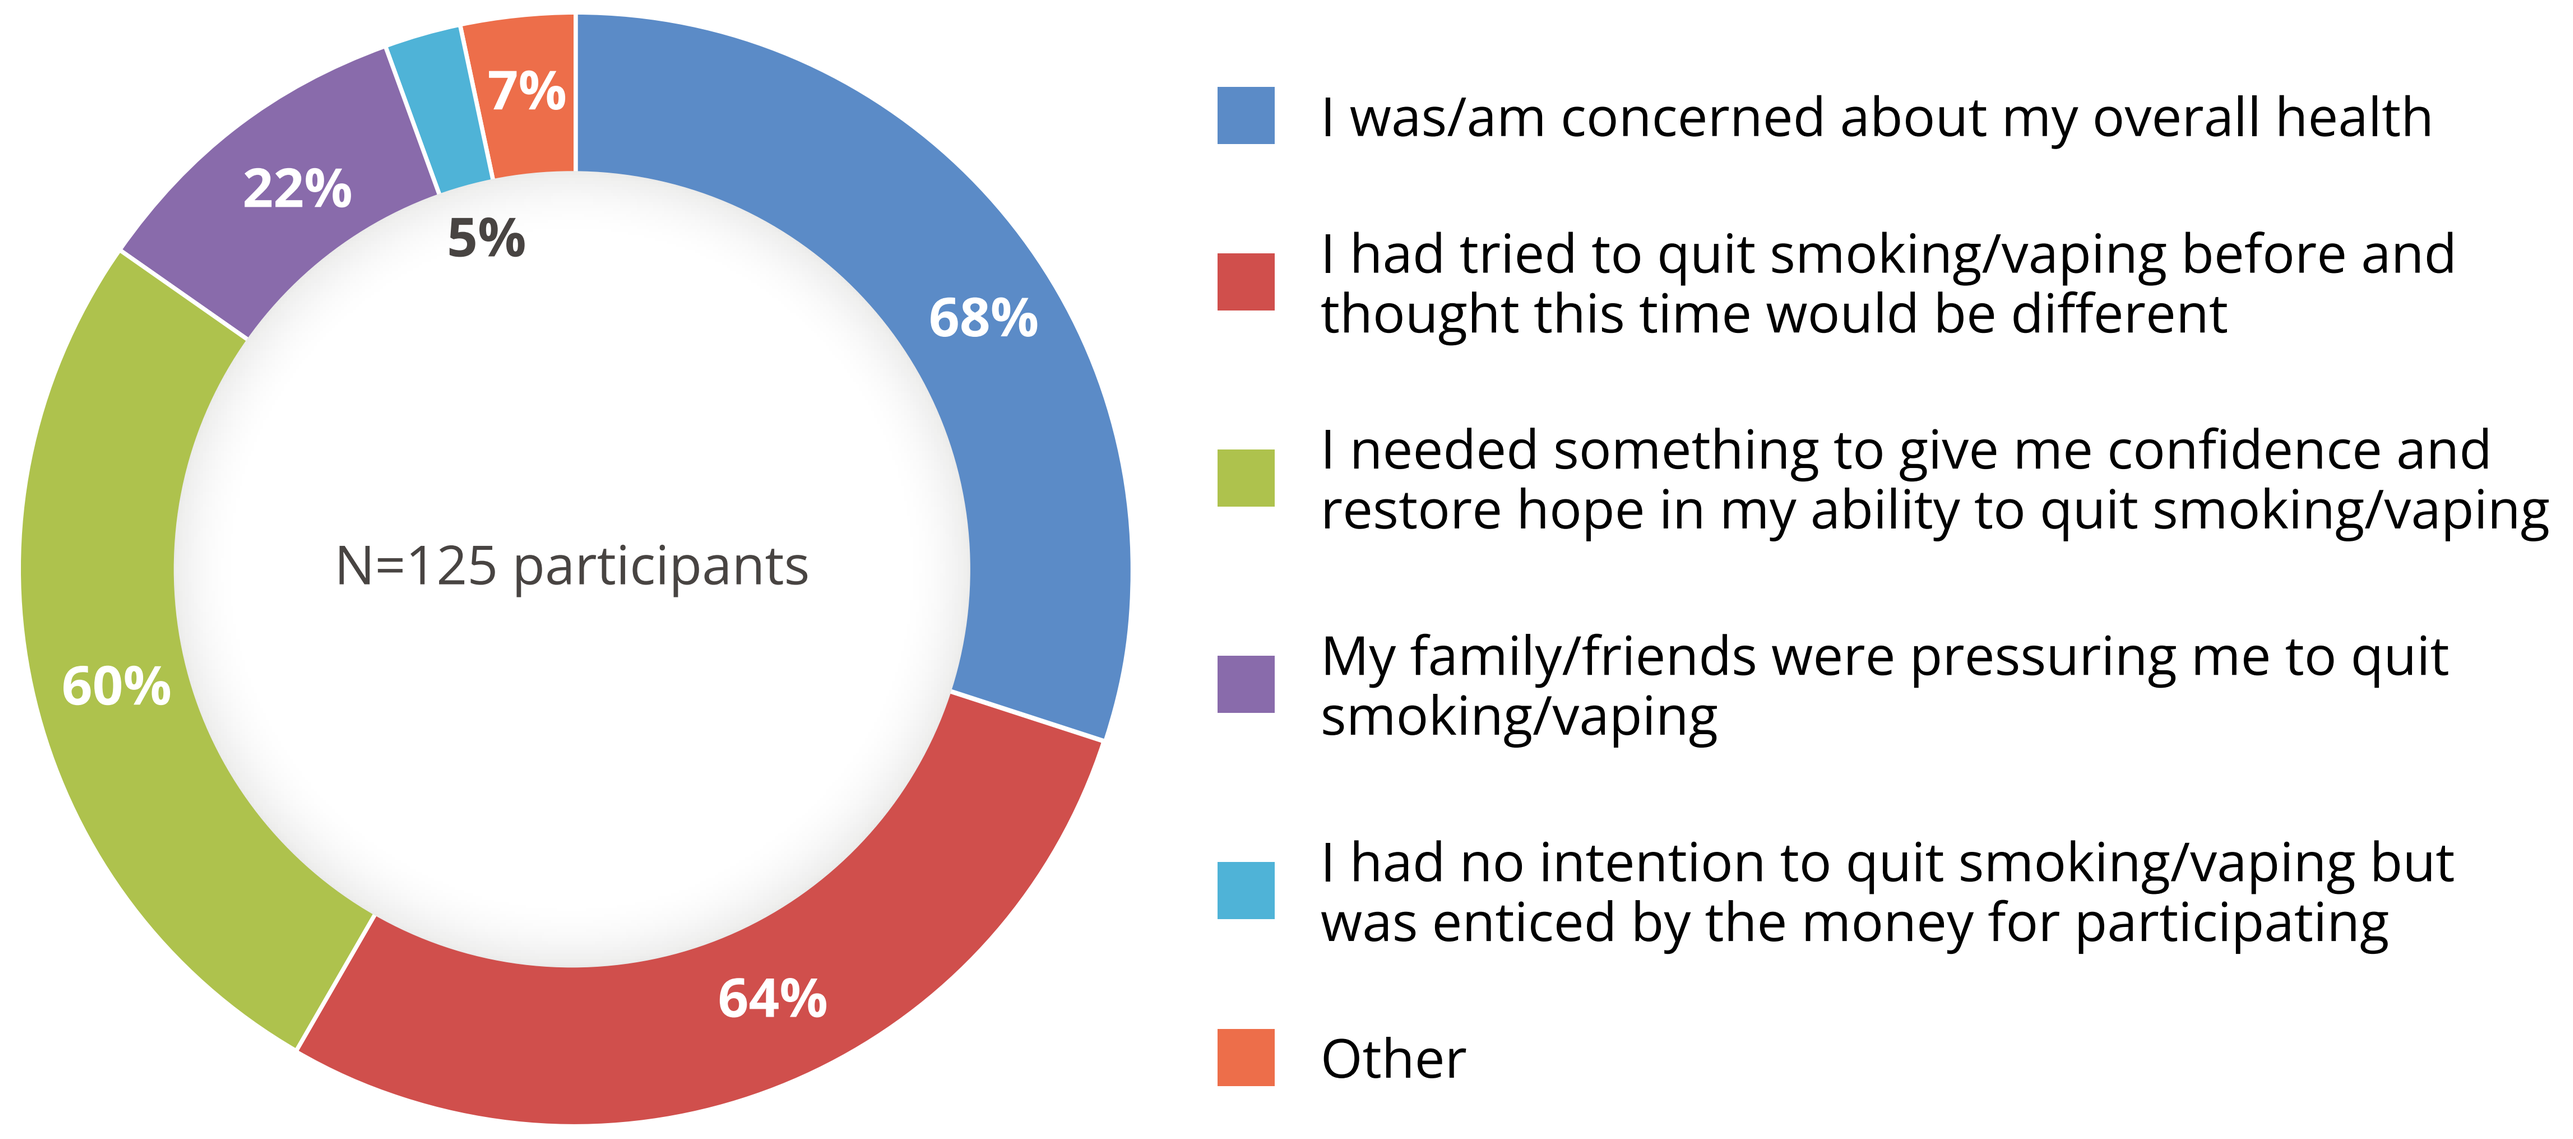

N=125 participants
68%
64%
60%
22%
5%
7%
I was/am concerned about my overall health
I had tried to quit smoking/vaping before and thought this time would be different
I needed something to give me confidence and restore hope in my ability to quit smoking/vaping
My family/friends were pressuring me to quit smoking/vaping
I had no intention to quit smoking/vaping but was enticed by the money for participating
Other

[a]Participants were permitted to select multiple responses.

B. Self-reported cessation

Did you completely quit smoking/vaping or reduce smoking/vaping while participating in the ORCA-OL study?

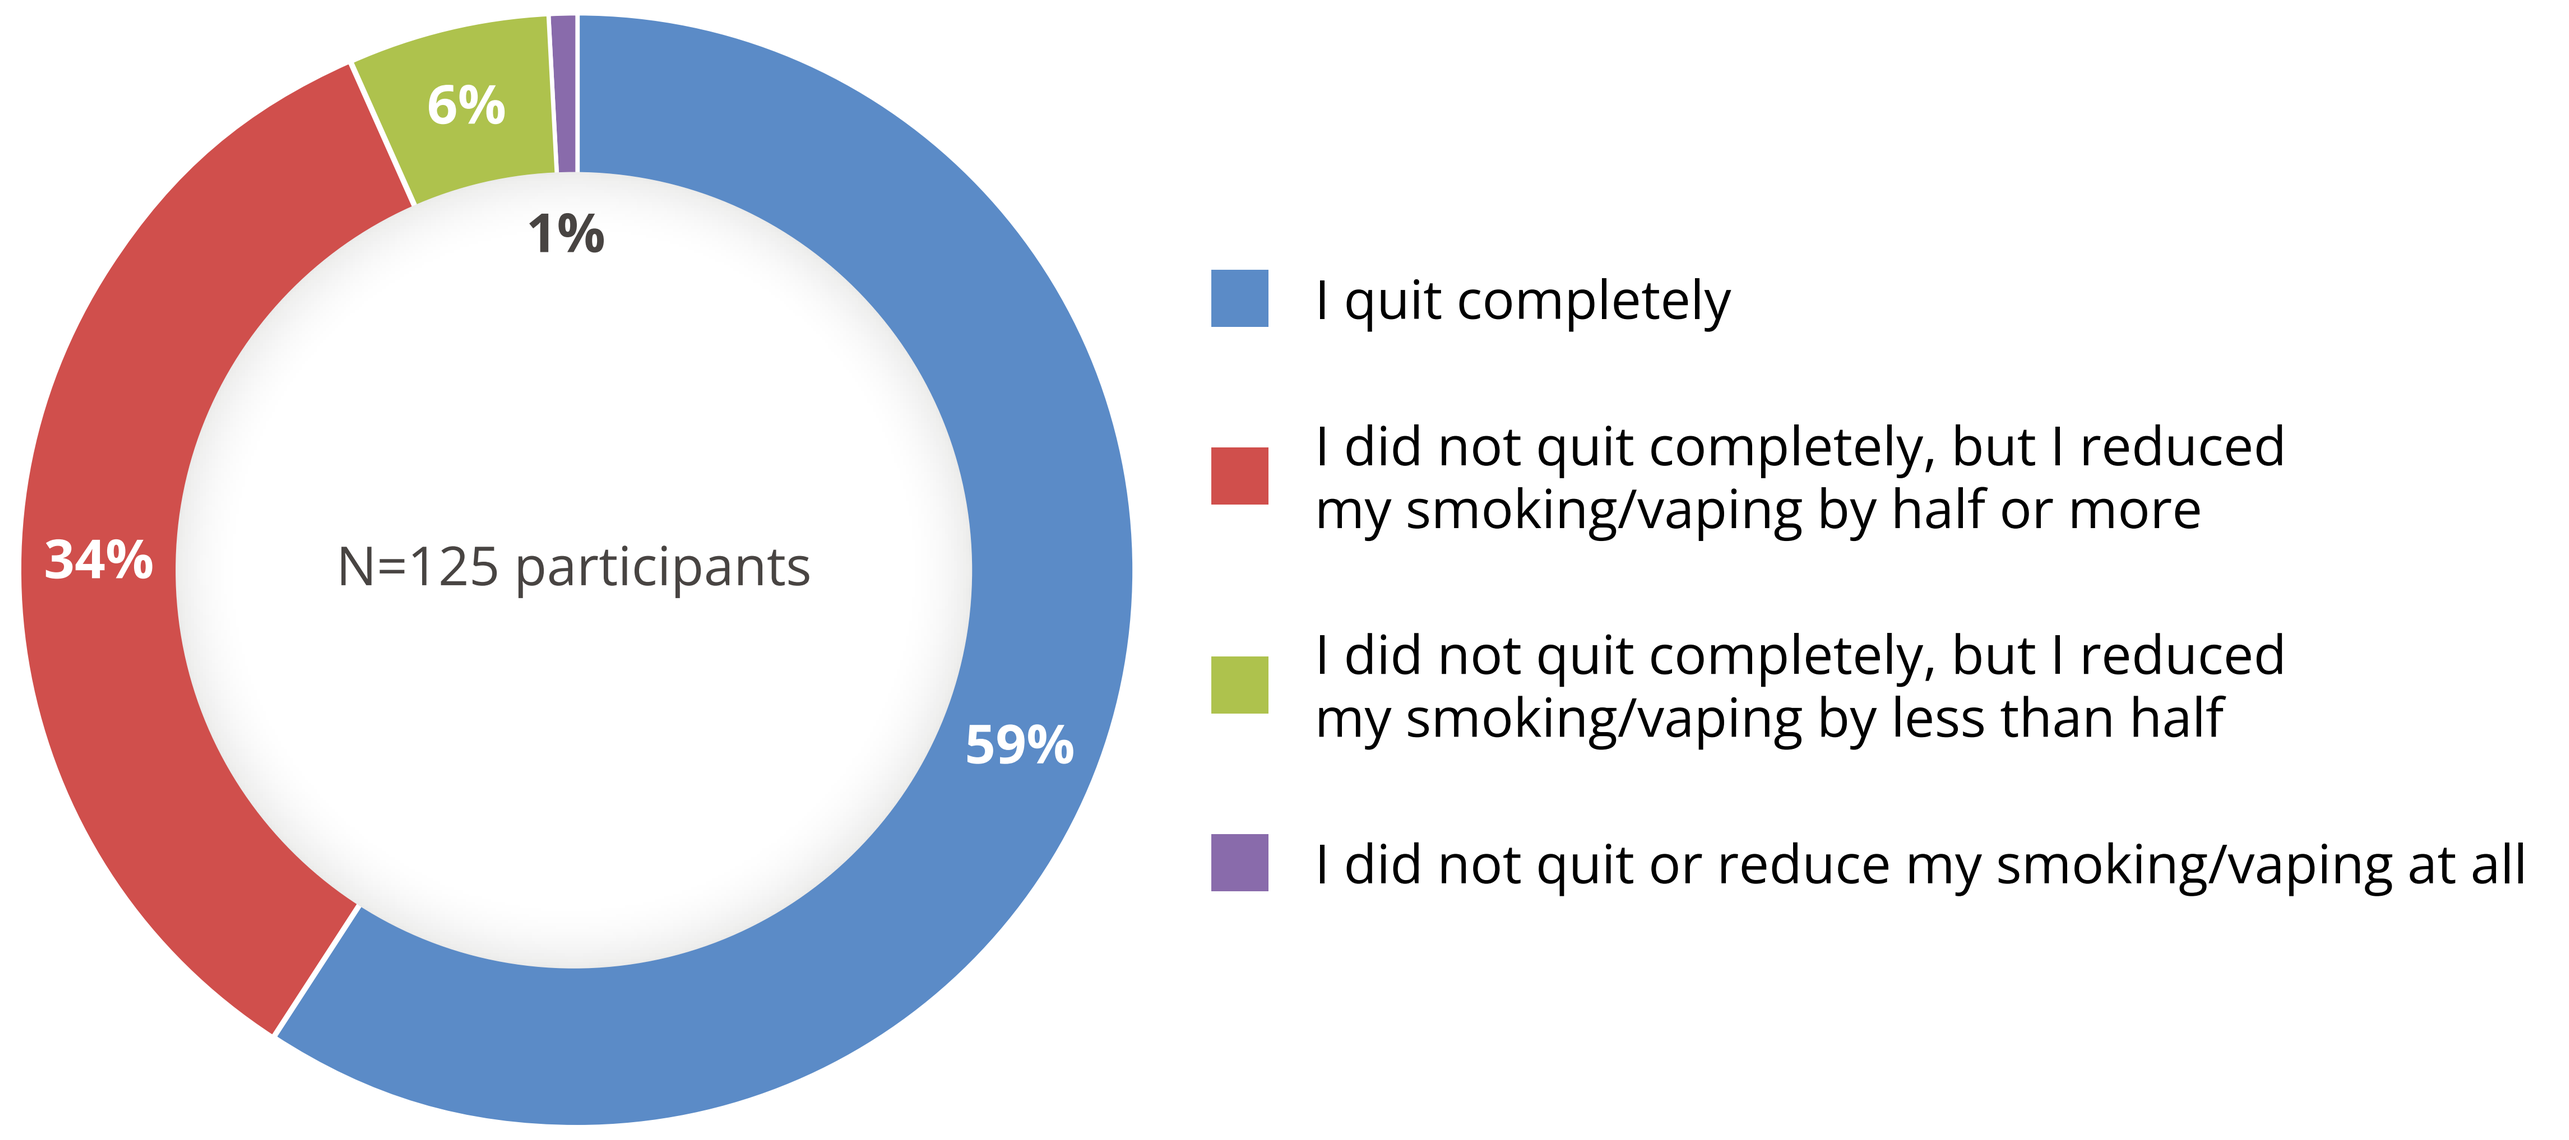

N=125 participants
59%
34%
6%
1%
I quit completely
I did not quit completely, but I reduced my smoking/vaping by half or more
I did not quit completely, but I reduced my smoking/vaping by less than half
I did not quit or reduce my smoking/vaping at all

C. Impressions of the impact of cytisinicline on their nicotine use

Do you believe the study medication contributed to your ability to stop or reduce your nicotine use?

Yes: 98% No: 2%

Please tell us why you selected your answer

"I have seen a dramatic reduction in my desire to smoke as well as no known side effects"

"Literally QUIT even thinking about having a cigarette. ZERO cravings"

"I did not experience any physical withdrawals, and when cigarettes are not available or I am in a non-smoking situation, I did not care … I attribute that to the medicine"

"It absolutely helped. It drastically reduced cravings. There are/were times when I would like to smoke but could easily resist"

"It helped give me the push to quit completely, I'm not sure if I could've done it without the medication"

"My vaping reduced. And continued to reduce. I started at 4-6 vapes per month. Now it's only 1"

Please select the reasons you believe the medication you received helped you to quit or reduce your smoking/vaping[a,b]

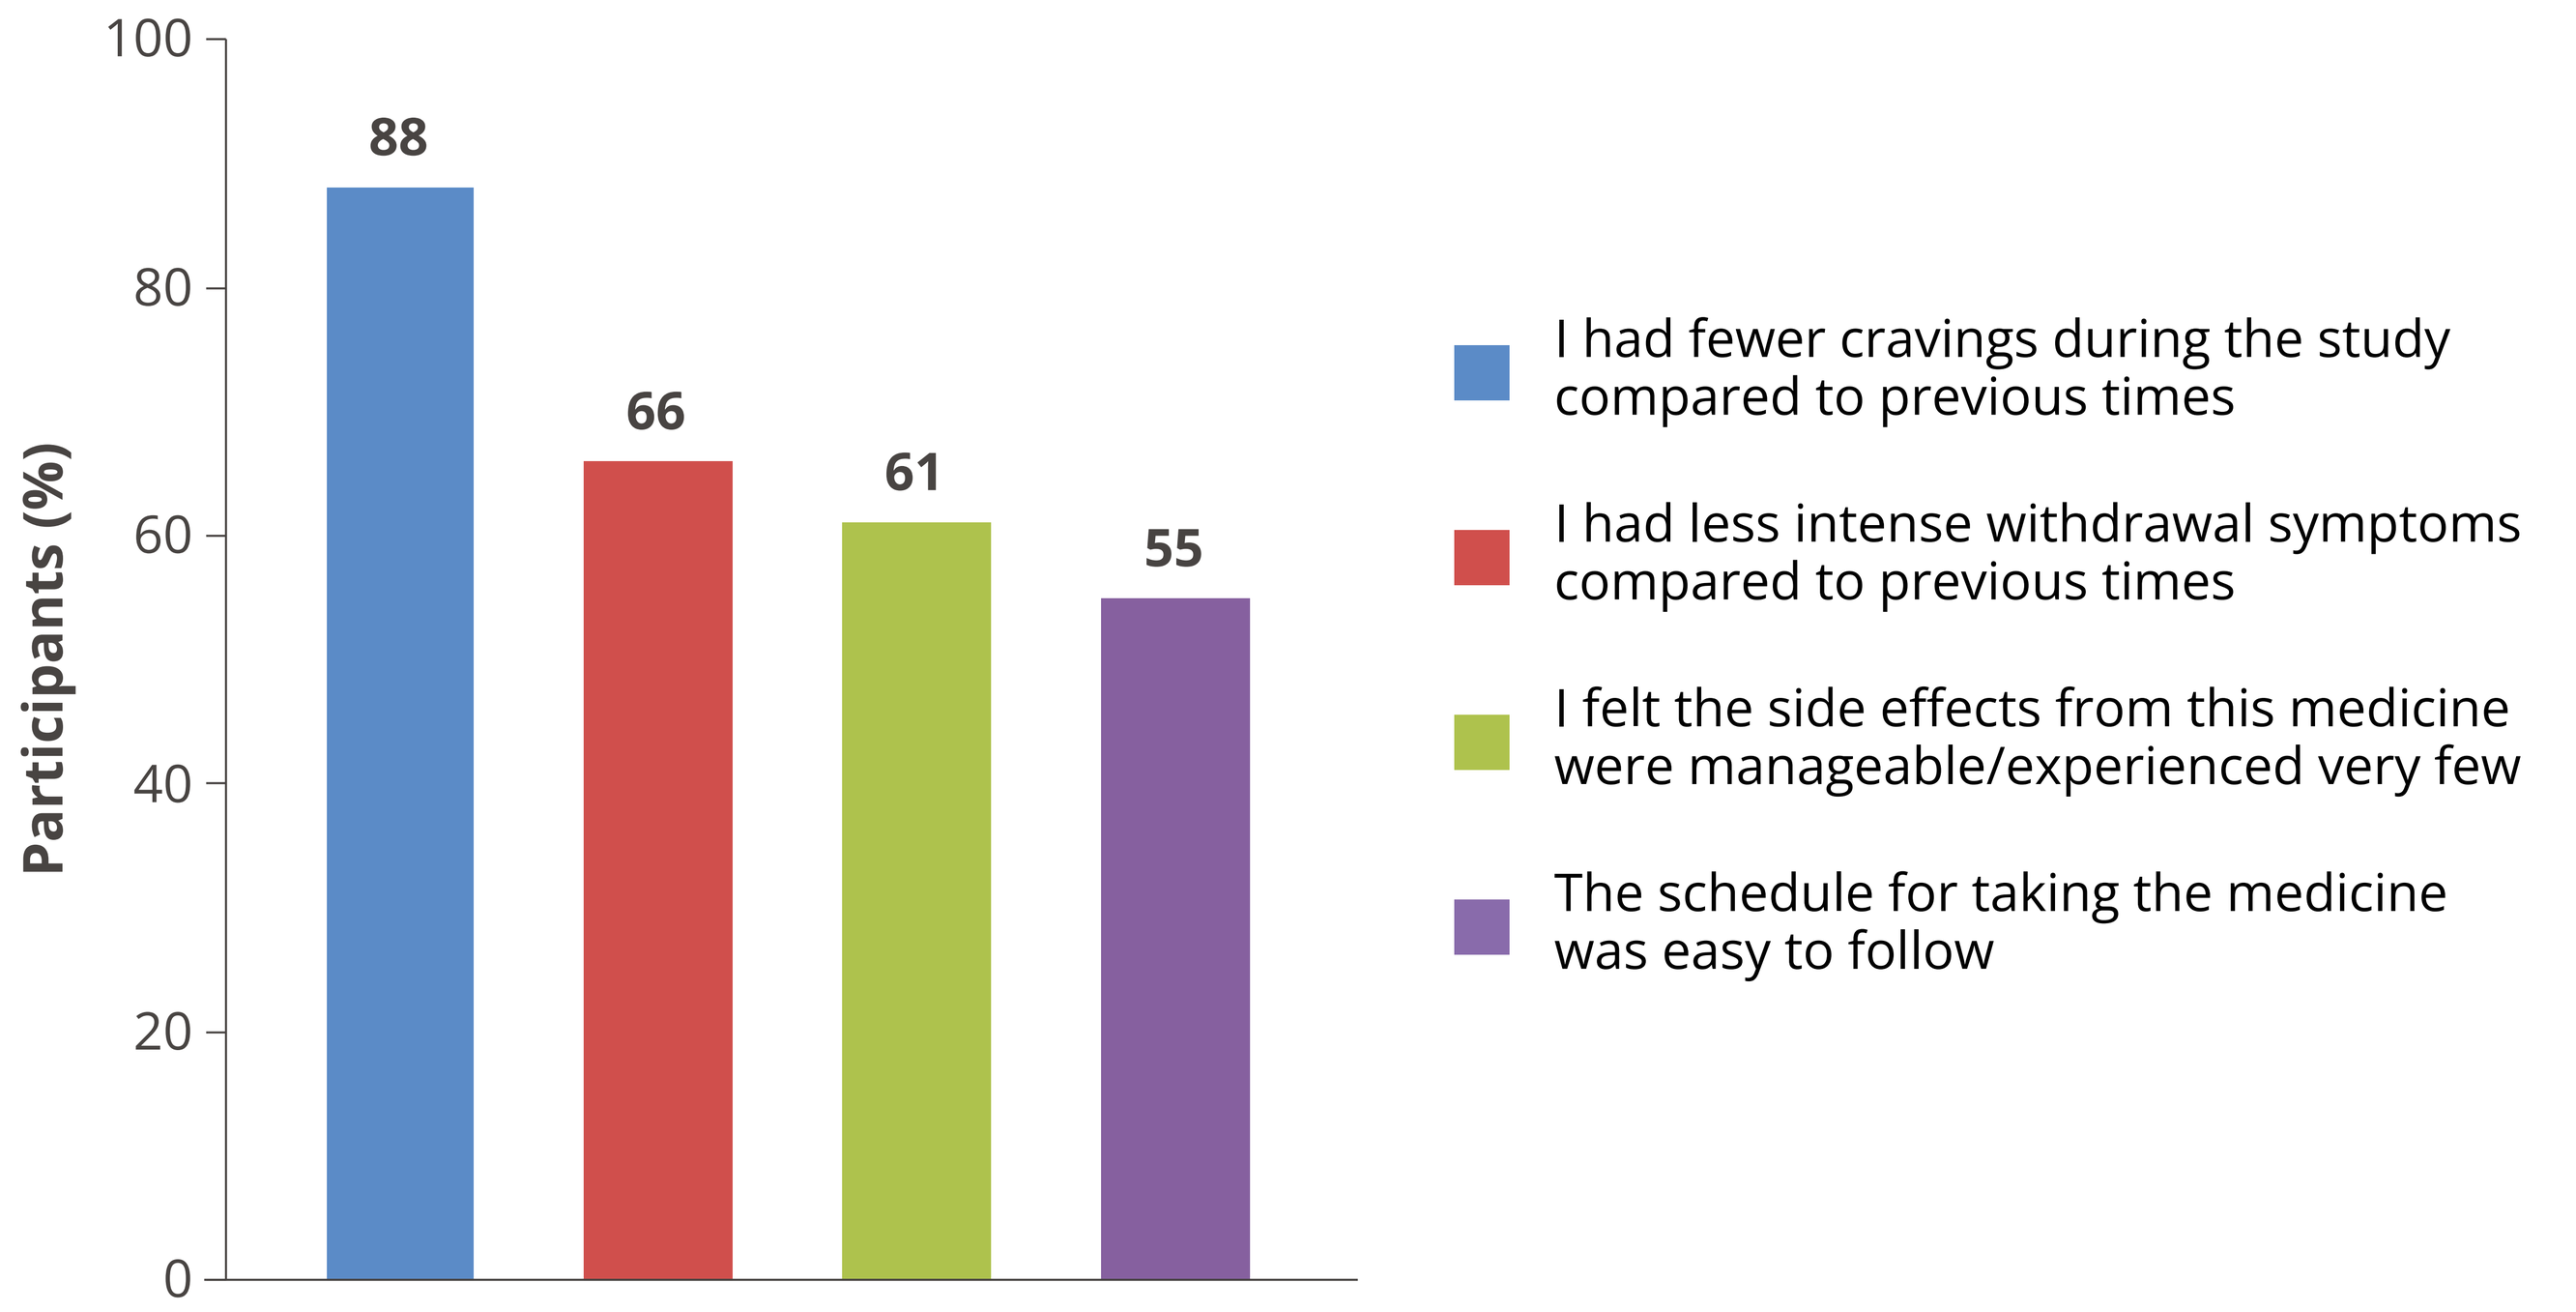

Participants (%)
100
80
60
40
20
0
88
66
61
55
I had fewer cravings during the study compared to previous times
I had less intense withdrawal symptoms compared to previous times
I felt the side effects from this medicine were manageable/experienced very few
The schedule for taking the medicine was easy to follow

[a]Participants who answered "yes" to question: Do you believe the study medication contributed to your ability to stop or reduce your nicotine use? [b]Participants were permitted to select multiple responses.

D. Benefits of quitting

If you have quit or reduced smoking/vaping since participating in the ORCA-OL trial, what do you feel are the biggest benefits you have seen or felt so far?[a]

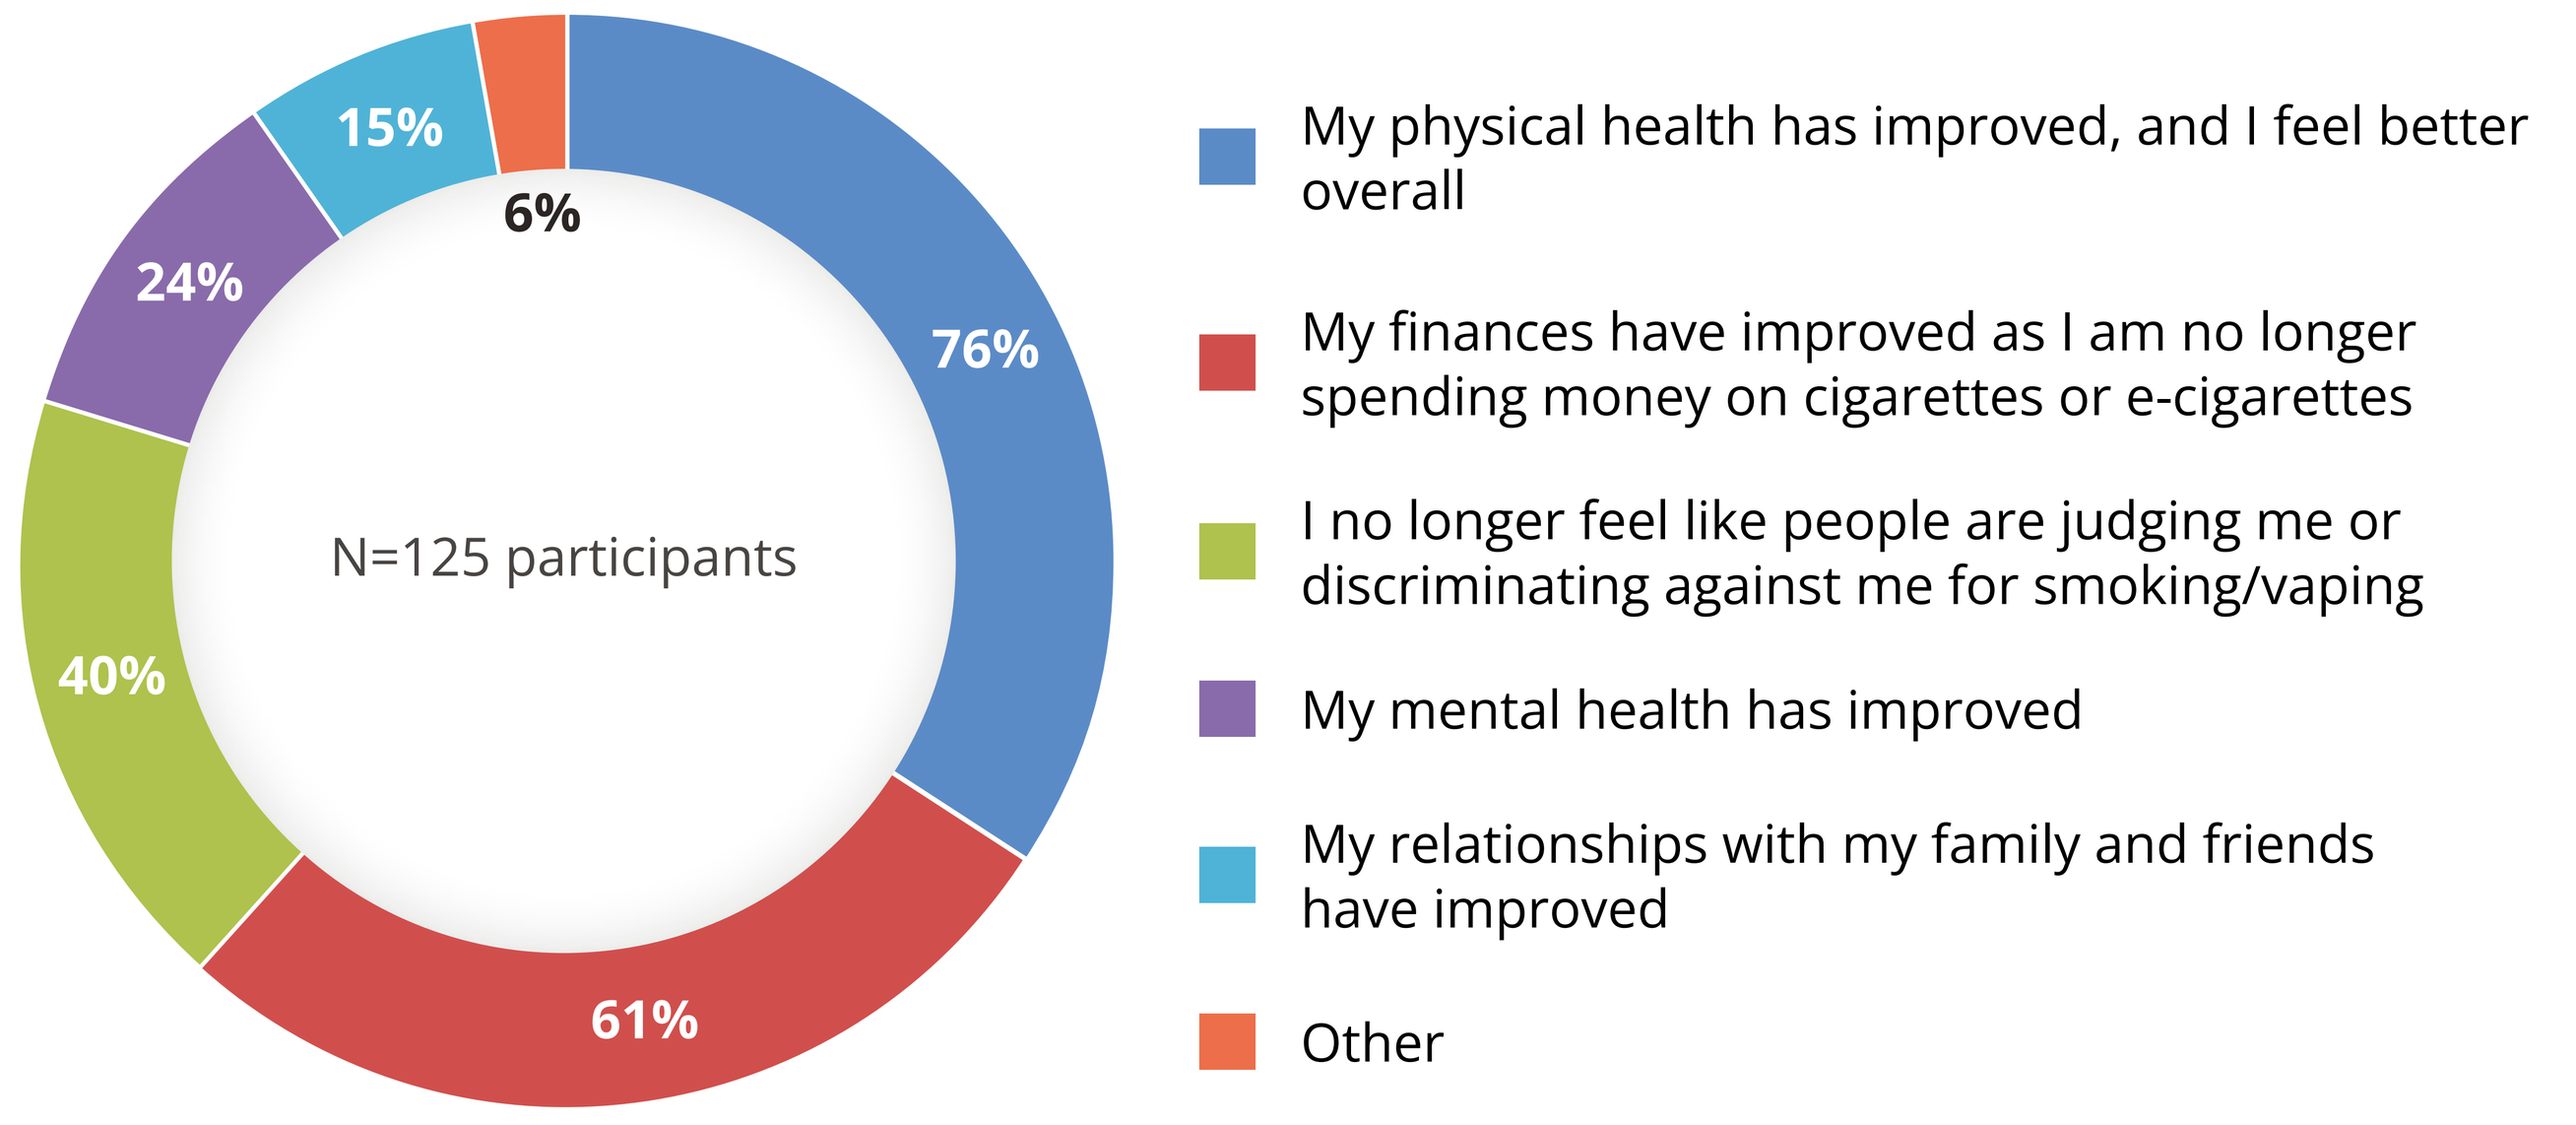

N=125 participants
76%
61%
40%
24%
15%
6%
My physical health has improved, and I feel better overall
My finances have improved as I am no longer spending money on cigarettes or e-cigarettes
I no longer feel like people are judging me or discriminating against me for smoking/vaping
My mental health has improved
My relationships with my family and friends have improved
Other

[a]Participants were permitted to select multiple responses.

Other survey findings

Overall, 98% of participants stated that they would recommend cytisinicline to a friend or family member trying to quit smoking/vaping

Discussion

- The ORCA-OL study evaluated the safety of long-term, 52-week cytisinicline exposure,[4] whereas the ORCA-2, ORCA-3, and ORCA-V1 studies assessed cytisinicline exposure over 6- and 12-week treatment durations.[1–3] The post-trial survey from ORCA-OL provides exploratory insights into participant experiences with extended cytisinicline use
- Survey participation was limited (125 of 475 ORCA-OL participants),[4] and as a voluntary, open-label survey, results may be biased toward individuals with more positive cessation experiences; self-reported outcomes should be interpreted with appropriate caution
- Findings from this small cohort of long-term exposure participants suggest favorable acceptability of cytisinicline and indicate potential for smoking and vaping cessation, warranting further study[4]

Conclusions

The findings from this voluntary post-trial survey of cytisinicline use during ORCA-OL showed that most participants reported:

- High quit and reduction rates of nicotine use, driven by fewer cravings
- Manageable or very few side effects
- Broad perceived benefits
- They would recommend cytisinicline to others

Acknowledgments: The authors acknowledge Julie Ball and Roxann Becco from Achieve Life Sciences (Seattle, WA, USA) for their contribution to the study. Medical writing support, under the guidance of the authors, was provided by Julie Gray, BSc, an employee from the Publications and Medical Affairs Division of Omnicom Health Medical Communications, and was funded by Achieve Life Sciences in accordance with Good Publication Practice, GPP 2022 (Ann Intern Med 2022;175:1298–304).

References: **1.** Rigotti NA et al. JAMA Intern Med. 2024;184:922–30; **2.** Rigotti NA et al. JAMA. 2023;330:152–60; **3.** Rigotti NA et al. JAMA Intern Med. 2025;185:648–55; **4.** Available at: https://clinicaltrials.gov/study/NCT06435221

Poster presented at the 32nd Society for Research on Nicotine and Tobacco (SRNT) Annual Meeting, March 4–7, 2026, Baltimore, MD, USA